Time-to-event model for acute exacerbations in idiopathic pulmonary fibrosis (IPF)

Fei Tang,[1,2] Benjamin Weber,[1] Susanne Stowasser,[3] Julia Korell[1]

[1]Translational Medicine & Clinical Pharmacology, Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, USA; [2]Department of Pharmaceutical Sciences, College of Pharmacy, University of Kentucky, Lexington, KY, USA;
[3]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany

PIII-070

Introduction

- IPF is a chronic disease of unknown cause characterized by progressive fibrosis of the lungs, affecting 10–60 people per 100,000 in the US.[1,2] Median survival is around 3.8 years in adults with IPF aged ≥65 years.[2]
- Each year, approximately 10–20% of patients with IPF experience acute exacerbations, which are associated with a high risk of mortality.[2]
- Nintedanib is one of two treatments approved for use in IPF.[1,3]
- Results from the two replicate Phase III, 52-week, randomized, double-blind, placebo-controlled INPULSIS trials demonstrated that nintedanib (150 mg BID orally) slowed disease progression but did not significantly affect overall prognosis or QoL.[4]
- Identifying risk factors to predict exacerbations is key to improving healthcare for people with IPF.

Aim

Using data from the INPULSIS Phase III trials (NCT01335464, NCT01335477),[4] this study aimed to identify exacerbation risk factors to inform development of a parametric time-to-event model that may be used to predict exacerbations in IPF.

Methods

Data-Set Generation

- A NONMEM 7.3-compatible INPULSIS data set was generated in R Studio (v1.1.383), with acute exacerbation as the endpoint of interest; censoring was applied at Day 372.
- Parametric survival analysis using NONMEM 7.3 was performed on time to first investigator-reported exacerbations, a key secondary endpoint in the INPULSIS trials.[4]

Model Development and Evaluation

- NONMEM v7.4.3 and PsN v4.8.1 software were used for development and evaluation. Different hazard functions (including exponential, Weibull, Gompertz and log-logistic distributions) were explored to develop the base model. Covariate models were developed using the following equations:

$$h(t) = h_0(t) \times \lambda_{cov}$$

where:

$h(t)$ = hazard at time t

$h_0(t)$ = base hazard at time t

$\lambda_{cov} = e^{\theta * cov}$ for binary categorical variables (cov = 0 or 1)

$\lambda_{cov} = e^{\theta * (cov - cov_{median})}$ for continuous variables

- A decrease of 3.84 in objective function value was the cutoff value (α=0.05) in univariate analysis.
- The final model was generated by performing stepwise covariate model selection on covariates significant in univariate analysis after removing highly correlated covariates with forward inclusion (P=0.05) and backward elimination (P=0.01).
- The model's goodness of fit was evaluated by visual predictive check with 100 simulations.

Results

Data-Set Overview

- Overall, 1,061 subjects were included in the analysis: 423 received placebo and 638 received 150 mg BID nintedanib.
- 69 investigator-reported acute exacerbations were recorded: 63 were a first event; 6 were a second event. Only time to first event was considered in this analysis (Figure 1).

Figure 1. Time to first acute exacerbation

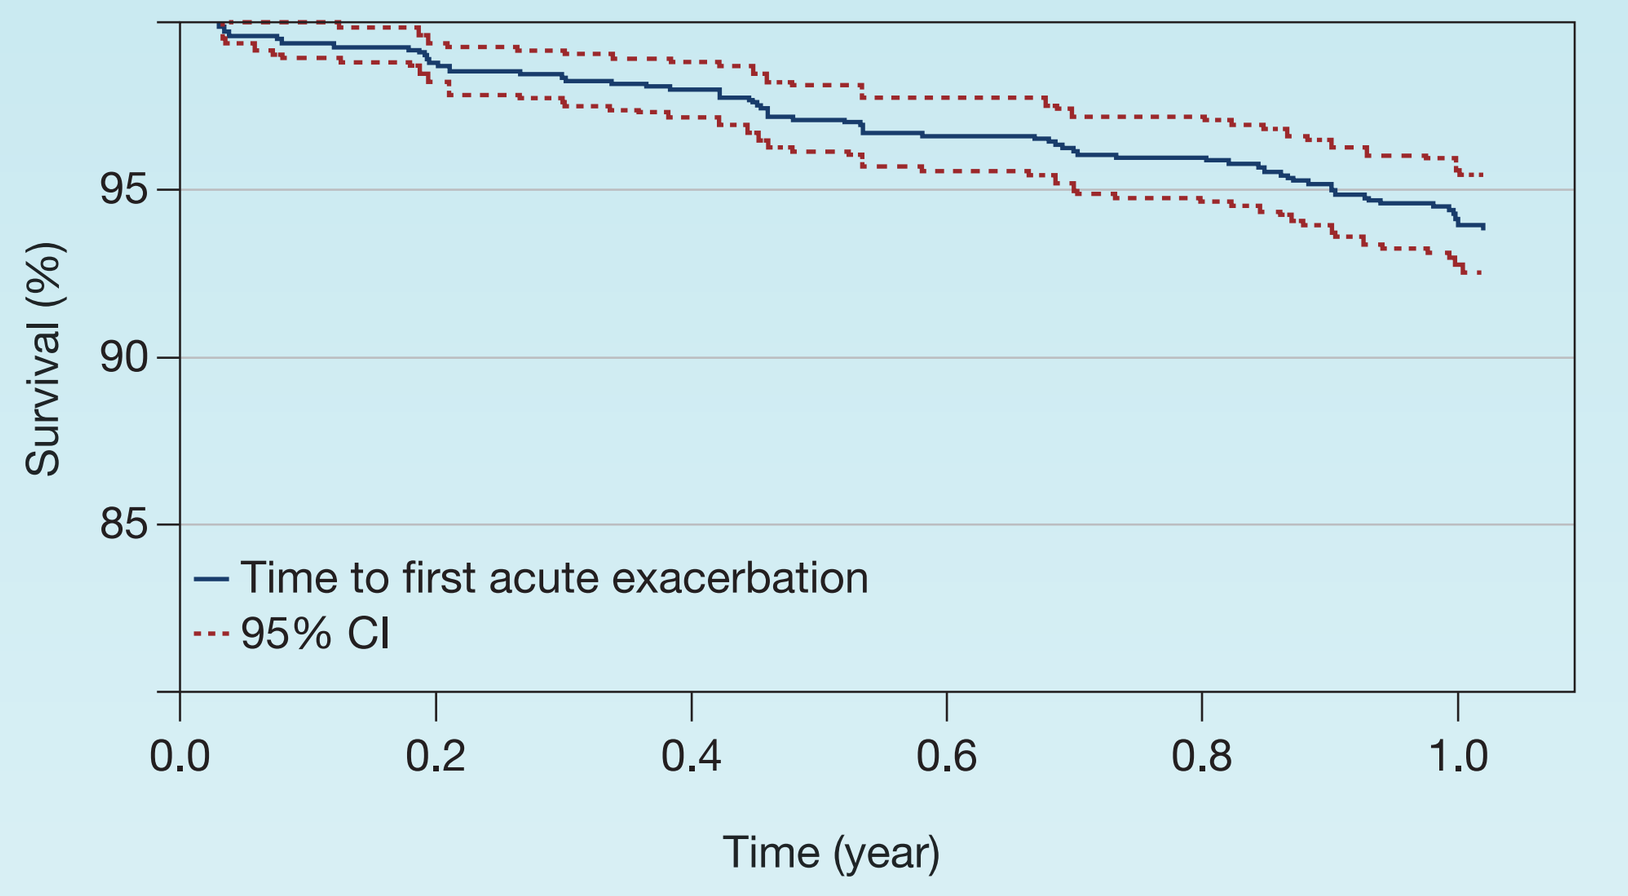
Survival (%)
95
90
85
Time to first acute exacerbation
95% CI
0.0
0.2
0.4
0.6
0.8
1.0
Time (year)

Structural Model

- Based on comparison of the objective function value, Akaike information criterion and standard error of the parameter estimates, the exponential model was used as the base model (Table 1).

Table 1. Parameter estimates of base hazard models considered

Parameter	Estimate	RSE (%)	AIC
Exponential model			479.966
λ (year $^{-1}$)	0.0612	12.6	
Weibull model			480.693
λ (year $^{-1}$)	0.0708	18.0	
γ	0.155	95.6	
Gompertz model			479.859
λ (year $^{-1}$)	0.0441	28.6	
γ	0.625	74.3	
Log-logistic model			480.851
λ (year $^{-1}$)	0.0632	13.0	
γ	1.17	12.8	

Univariate Analysis

Table 2. Variables included in univariate analysis

Demographics	Treatment-related	Disease-related
Age	Treatment vs placebo	Observed baseline FVC (L)
Sex	Nintedanib dose	Observed baseline % predicted FVC
Height	Systemic corticosteroids	Baseline % predicted DL_{CO}
BMI	Bronchodilators	Presence of emphysema
BSA	Supplemental oxygen	Slopes of FVC decline[a]
Race	PPI or H_2I	Slopes of % predicted FVC decline[a]
Smoking	-	Δ $FVC_{(baseline \rightarrow Week\ 52)}$ (L)[a]
Alcohol use	-	Δ % predicted $FVC_{(baseline \rightarrow Week\ 52)}$[a]

[a]Derived from model developed from nintedanib Phase II and Phase III studies.

Figure 2. Forest plot summarizing results from univariate analysis

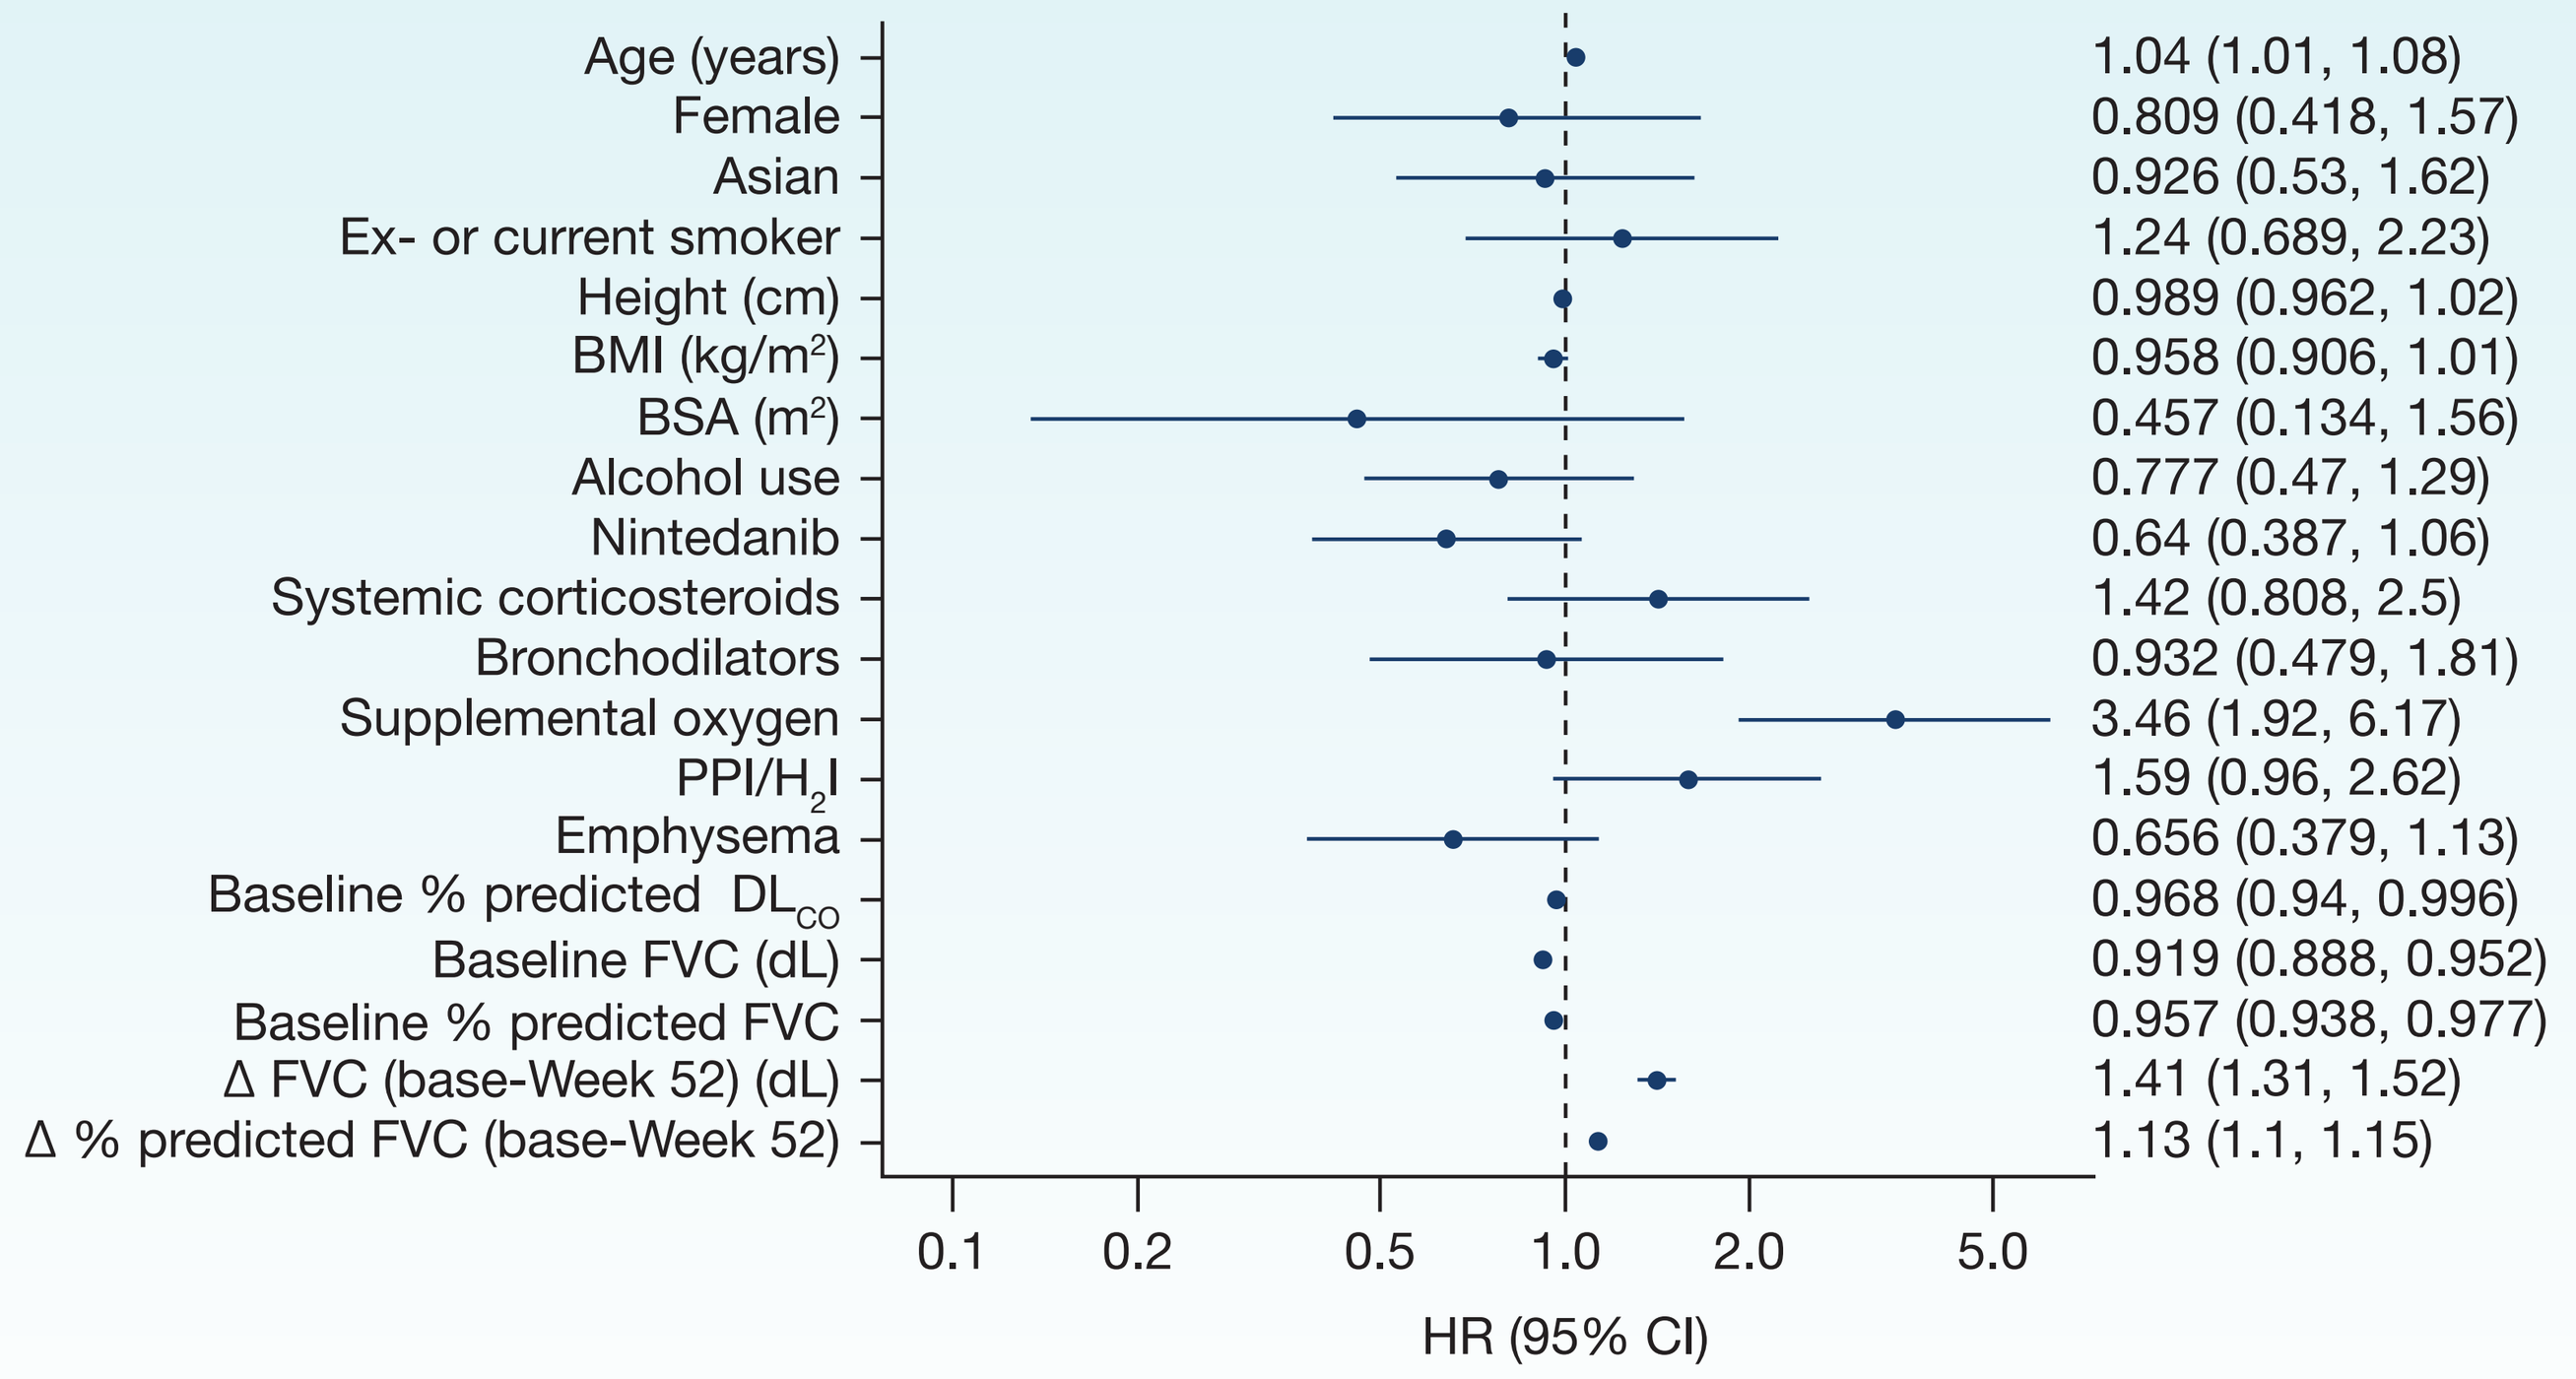
Age (years) 1.04 (1.01, 1.08)
Female 0.809 (0.418, 1.57)
Asian 0.926 (0.53, 1.62)
Ex- or current smoker 1.24 (0.689, 2.23)
Height (cm) 0.989 (0.962, 1.02)
BMI (kg/m²) 0.958 (0.906, 1.01)
BSA (m²) 0.457 (0.134, 1.56)
Alcohol use 0.777 (0.47, 1.29)
Nintedanib 0.64 (0.387, 1.06)
Systemic corticosteroids 1.42 (0.808, 2.5)
Bronchodilators 0.932 (0.479, 1.81)
Supplemental oxygen 3.46 (1.92, 6.17)
PPI/H2I 1.59 (0.96, 2.62)
Emphysema 0.656 (0.379, 1.13)
Baseline % predicted DLCO 0.968 (0.94, 0.996)
Baseline FVC (dL) 0.919 (0.888, 0.952)
Baseline % predicted FVC 0.957 (0.938, 0.977)
Δ FVC (base-Week 52) (dL) 1.41 (1.31, 1.52)
Δ % predicted FVC (base-Week 52) 1.13 (1.1, 1.15)
0.1
0.2
0.5
1.0
2.0
5.0
HR (95% CI)

Multivariate Analysis and Model Evaluation

- Variables included in stepwise covariate model:
 - baseline age
 - observed baseline % predicted FVC
 - baseline % predicted DL_{CO}
 - supplemental oxygen use
 - change in model-predicted $FVC_{(baseline \rightarrow Week\ 52)}$.
- Variables retained in the final model after stepwise covariate model analysis (Table 3):
 - baseline age
 - supplemental oxygen use
 - observed baseline % predicted FVC
 - change in model-predicted $FVC_{(baseline \rightarrow Week\ 52)}$.
- Objective function value of the final model = 378.511

Table 3. Parameter estimates of the final covariate model

Parameter (on λ)	Estimate	RSE (%)
λ (year $^{-1}$)	0.0300	17.8
Age	0.0474	36.2
Supplemental oxygen	1.06	28.7
Change in model predicted $FVC_{(baseline \rightarrow Week\ 52)}$	3.69	12.3
Observed baseline % predicted FVC	–0.0366	28.5

Figure 3. Overall and stratified visual predictive check of the final covariate model

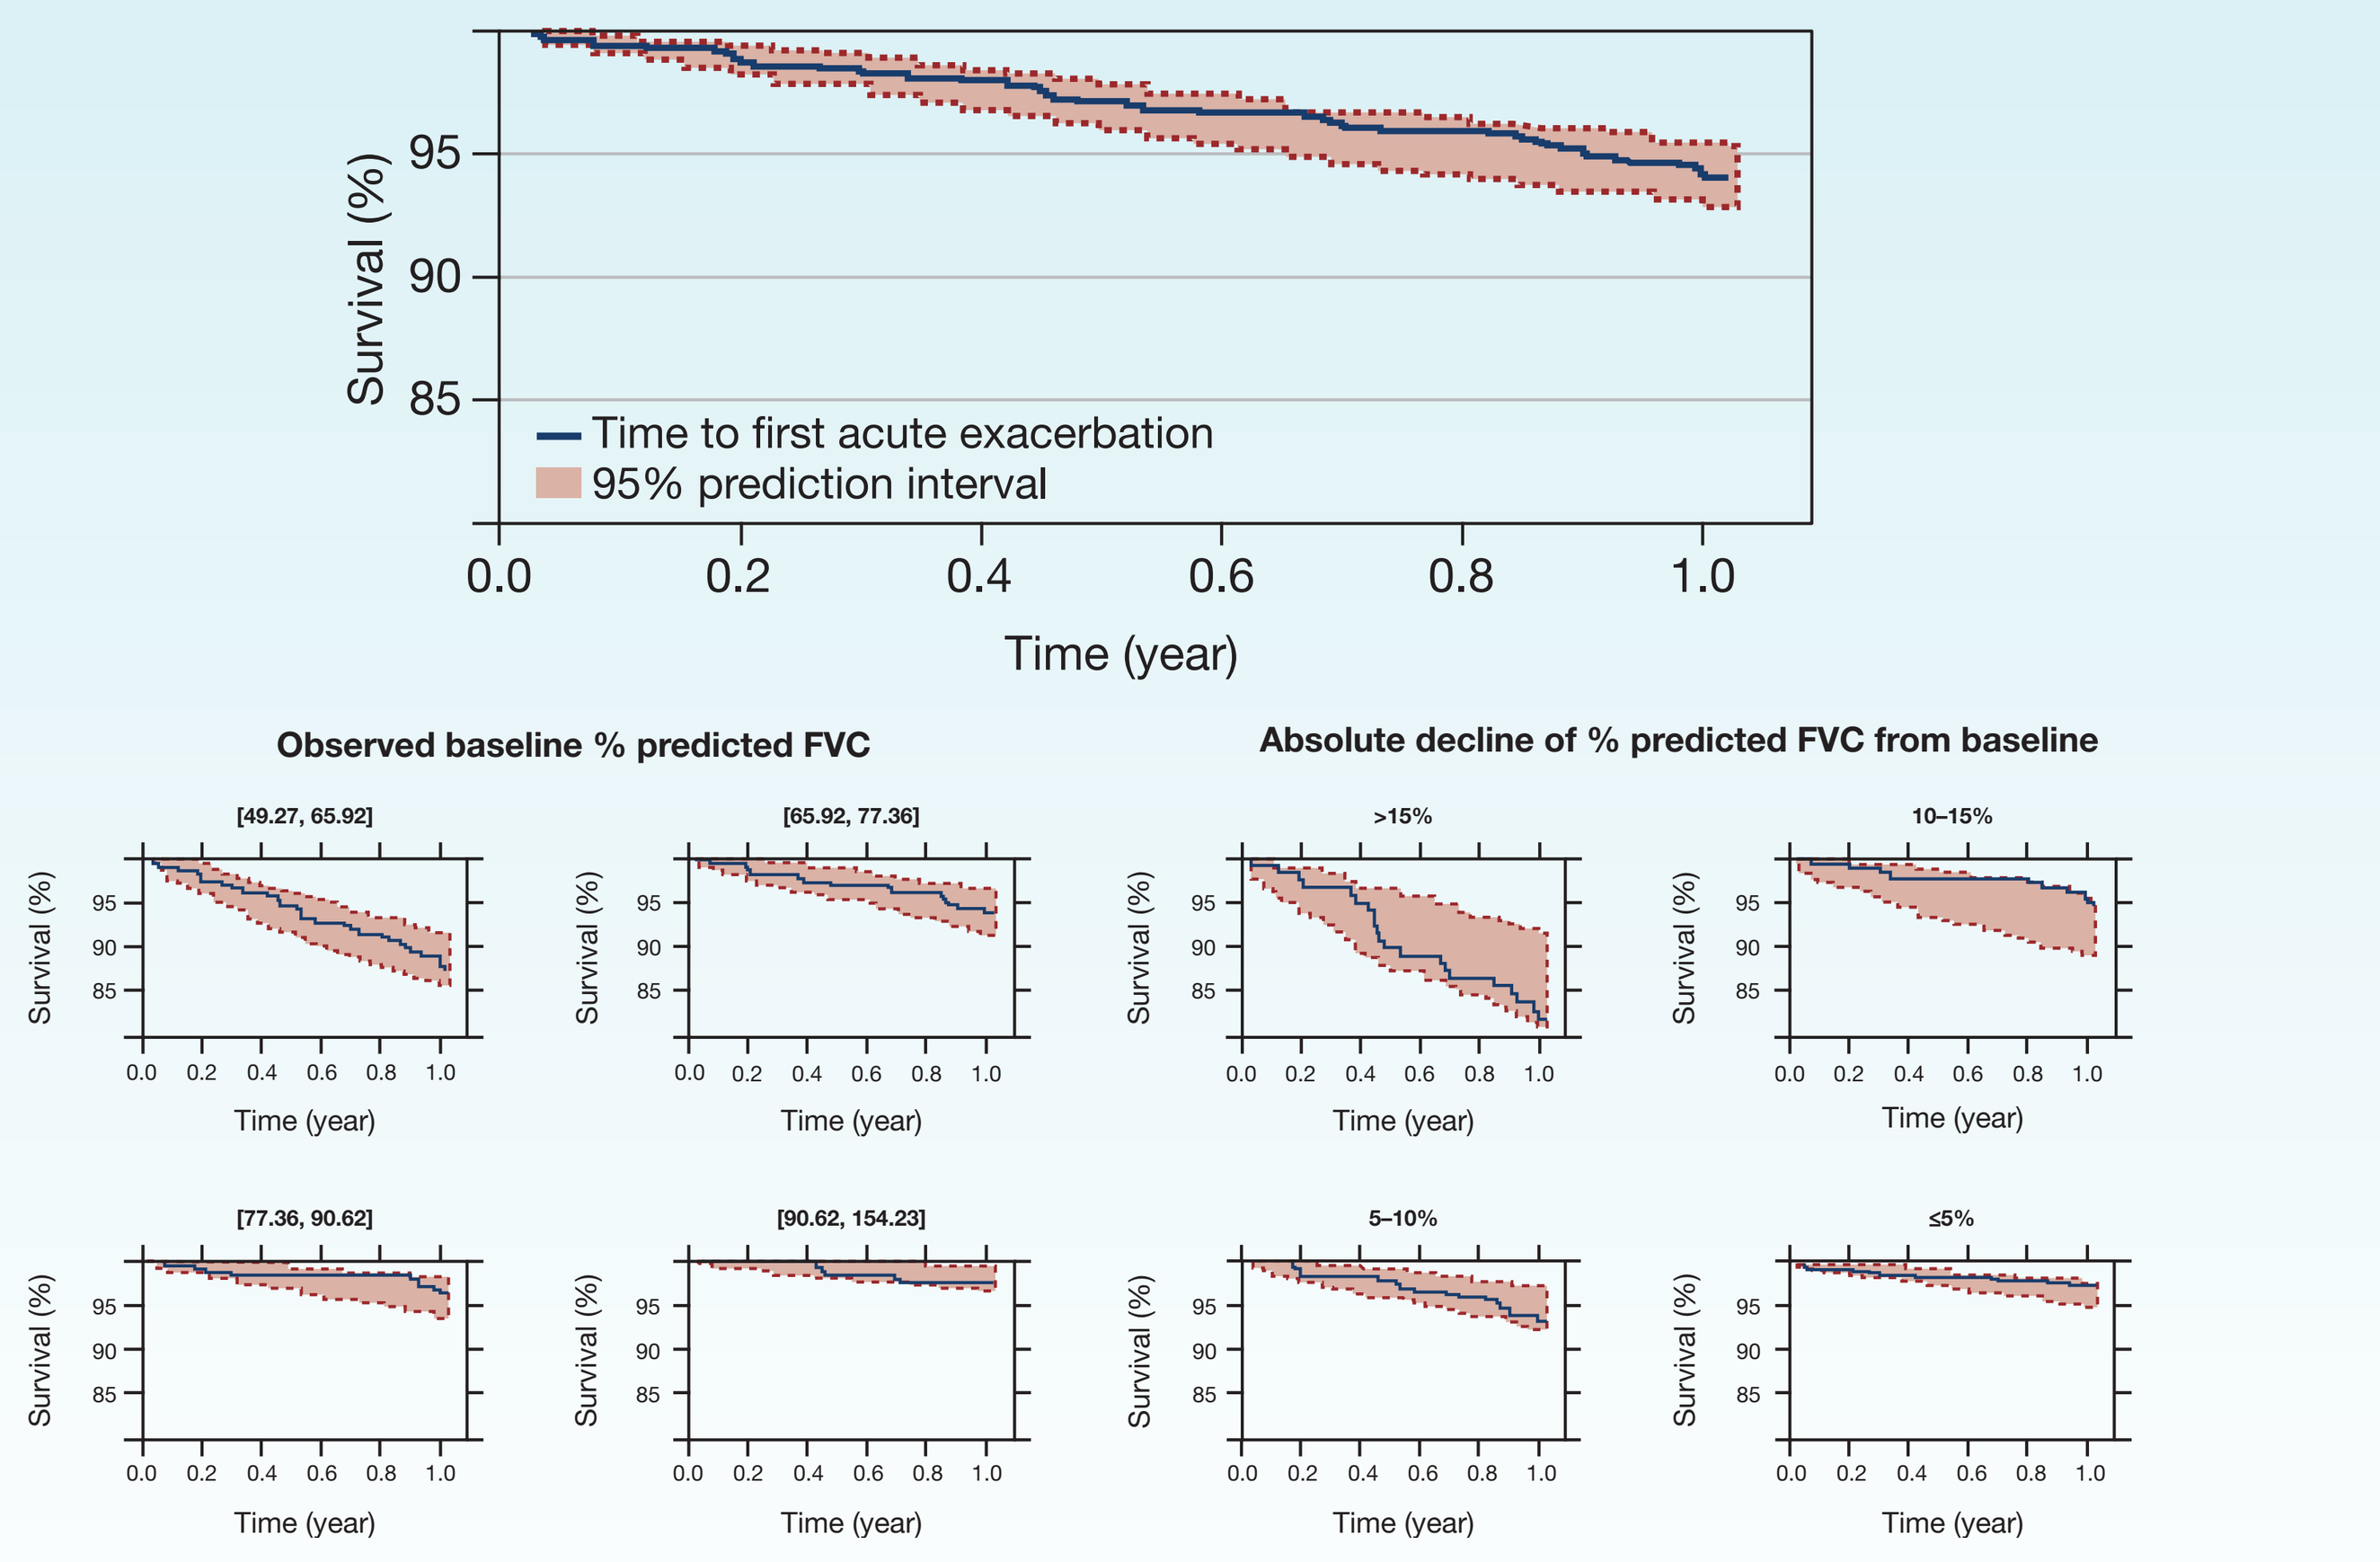
Survival (%)
95
90
85
Time to first acute exacerbation
95% prediction interval
0.0
0.2
0.4
0.6
0.8
1.0
Time (year)
Observed baseline % predicted FVC
Absolute decline of % predicted FVC from baseline
[49.27, 65.92]
[65.92, 77.36]
>15%
10–15%
[77.36, 90.62]
[90.62, 154.23]
5–10%
≤5%

Discussion

- A parametric time-to-event model was developed for acute exacerbations in IPF.
- Following stepwise covariate model analysis, significant covariates predictive of exacerbations were identified, including:
 - baseline age
 - supplemental oxygen use
 - baseline % predicted FVC
 - decline in FVC over 52 weeks.
- Overall and stratified visual predictive check plots demonstrated adequate predictive performance of the model (Figure 3).
- Supplemental oxygen use served as a proxy for disease severity at baseline and was incorporated in the final time-to-event model.
- Dropout modeling was not performed due to reporting bias arising from operational study procedures.
- Due to low number of second events, a repeated time-to-event analysis was not possible.

Conclusions

The developed parametric time-to-event model is a first step towards predicting exacerbations in IPF.

This model has the potential to improve healthcare for people with IPF by facilitating earlier identification of those at risk of exacerbations.

References

1. Martinez FJ, et al. Nat Rev Dis Primers 2017; 3:17074;
2. Lederer DJ, Martinez FJ. N Engl J Med 2018; 378(19):1811–1823;
3. Raghu G, et al. Am J Respir Crit Care Med 2015; 192(2):e3–e19;
4. Richeldi L, et al. N Engl J Med 2014; 370:2071–2082.

Acknowledgments

This study was supported by Boehringer Ingelheim. Caroline Sproat at MediTech Media provided editorial assistance in the development of this poster, funded by Boehringer Ingelheim. The authors would like to thank all the patients and their families for participating in the INPULSIS trials, and extend their thanks to the investigators.

Disclosures

FT reports personal fees from Boehringer Ingelheim. BW, SS and JK are paid employees of Boehringer Ingelheim.

Abbreviations

λ, base hazard; AIC, Akaike information criterion; BID, twice daily; BMI, body mass index; BSA, body surface area; CI, confidence interval; DL_{CO}, diffusing capacity of the lung for carbon monoxide; FVC, forced vital capacity; H_2I, histamine inhibitor; HR, hazard ratio; IPF, idiopathic pulmonary fibrosis; PPI, proton pump inhibitor; QoL, quality of life; RSE, relative standard error.

Poster presented at the American Society for Clinical Pharmacology & Therapeutics Annual Meeting, March 13–16, 2019, Washington DC, USA .